Poster nr. 502

Recommended phase 2 dose, pharmacokinetics, pharmacodynamics, and preliminary efficacy of the IL-15 superagonist SO-C101 as monotherapy in patients with advanced/metastatic solid tumours

Stephane Champiat[1], Aurelien Marabelle[1], Vladimir Galvao[2], Aung Naing[3], Filip Janku[3], Patricia LoRusso[4], Peter Grell[5], Lenka Palova Jelinkova[7], Irena Adkins[7], Ulrich Moebius[7], Richard Sachse[6], David Bechard[8], Joachim Kiemle-Kallee[6], Ellena Garralda[2];

SC103 Investigators: 1. Institute Gustave Roussy Cancerologie, Villejuif, France; 2.Hospital Universitari Vall d'Hebron, Institute of Oncology (VHIO), Barcelona, Spain; 3. Department of Investigational Cancer Therapeutics, MD Anderson Cancer Center, Houston, TX; 4. Yale School of Medicine, New Haven, CT, United States; 5. Masaryk Memorial Cancer Institute, Brno, Czech Republic; 6. SOTIO Biotech AG, Basel, Switzerland; 7. SOTIO Biotech a.s, Prague, Czech Republic; 8. Cytune Pharma S.A.S., Nantes, France;

Introduction

- SO-C101 (future compound code: SOT101) is a superagonist fusion protein of IL-15 and the IL-15 receptor α sushi+ domain that selectively activates and expands natural killer (NK) and cytotoxic ($CD8^+$) T cells (1, 2).
- SO-C101 was shown to bind to the intermediate affinity IL-2/15Rβγ with high affinity and specificity (1, 2). It also exhibits enhanced pharmacokinetic and selective pharmacodynamics properties with improved antitumor efficacy relative to IL-2 and IL-15 in preclinical studies (3, 4).
- In this phase 1/1b multicentre, open-label, dose escalation study SO-C101 monotherapy was investigated in patients with selected advanced/metastatic tumours (NCT04234113).
- In addition, an individual case study is shown.

Methods

- Study objectives were to determine the maximum tolerated dose (MTD), the recommended phase 2 dose (RP2D), pharmacokinetics, pharmacodynamics, and preliminary efficacy of SO-C101 as monotherapy in patients with advanced/metastatic solid tumours.
- SO-C101 was administered as s.c. injection on day 1, 2, 8 and 9 of each 21 day cycle.
- The evaluation period for dose-limiting toxicities was 21 days.
- The RP2D was defined as MTD or below, also considering pharmacokinetic and pharmacodynamic parameters.
- Efficacy was evaluated according to RECiST 1.1 and iRECiST.
- After progression on SO-C101 monotherapy crossover to SO-C101 + pembrolizumab combination therapy was allowed.

Patient population

- Thirty patients with a median of 3 (range 1-9) lines of previous systemic therapies were treated at dose levels 0.25, 0.75, 1.5, 3.0, 6.0., 9.0, 12.0, and 15 µg/kg BW.

Safety

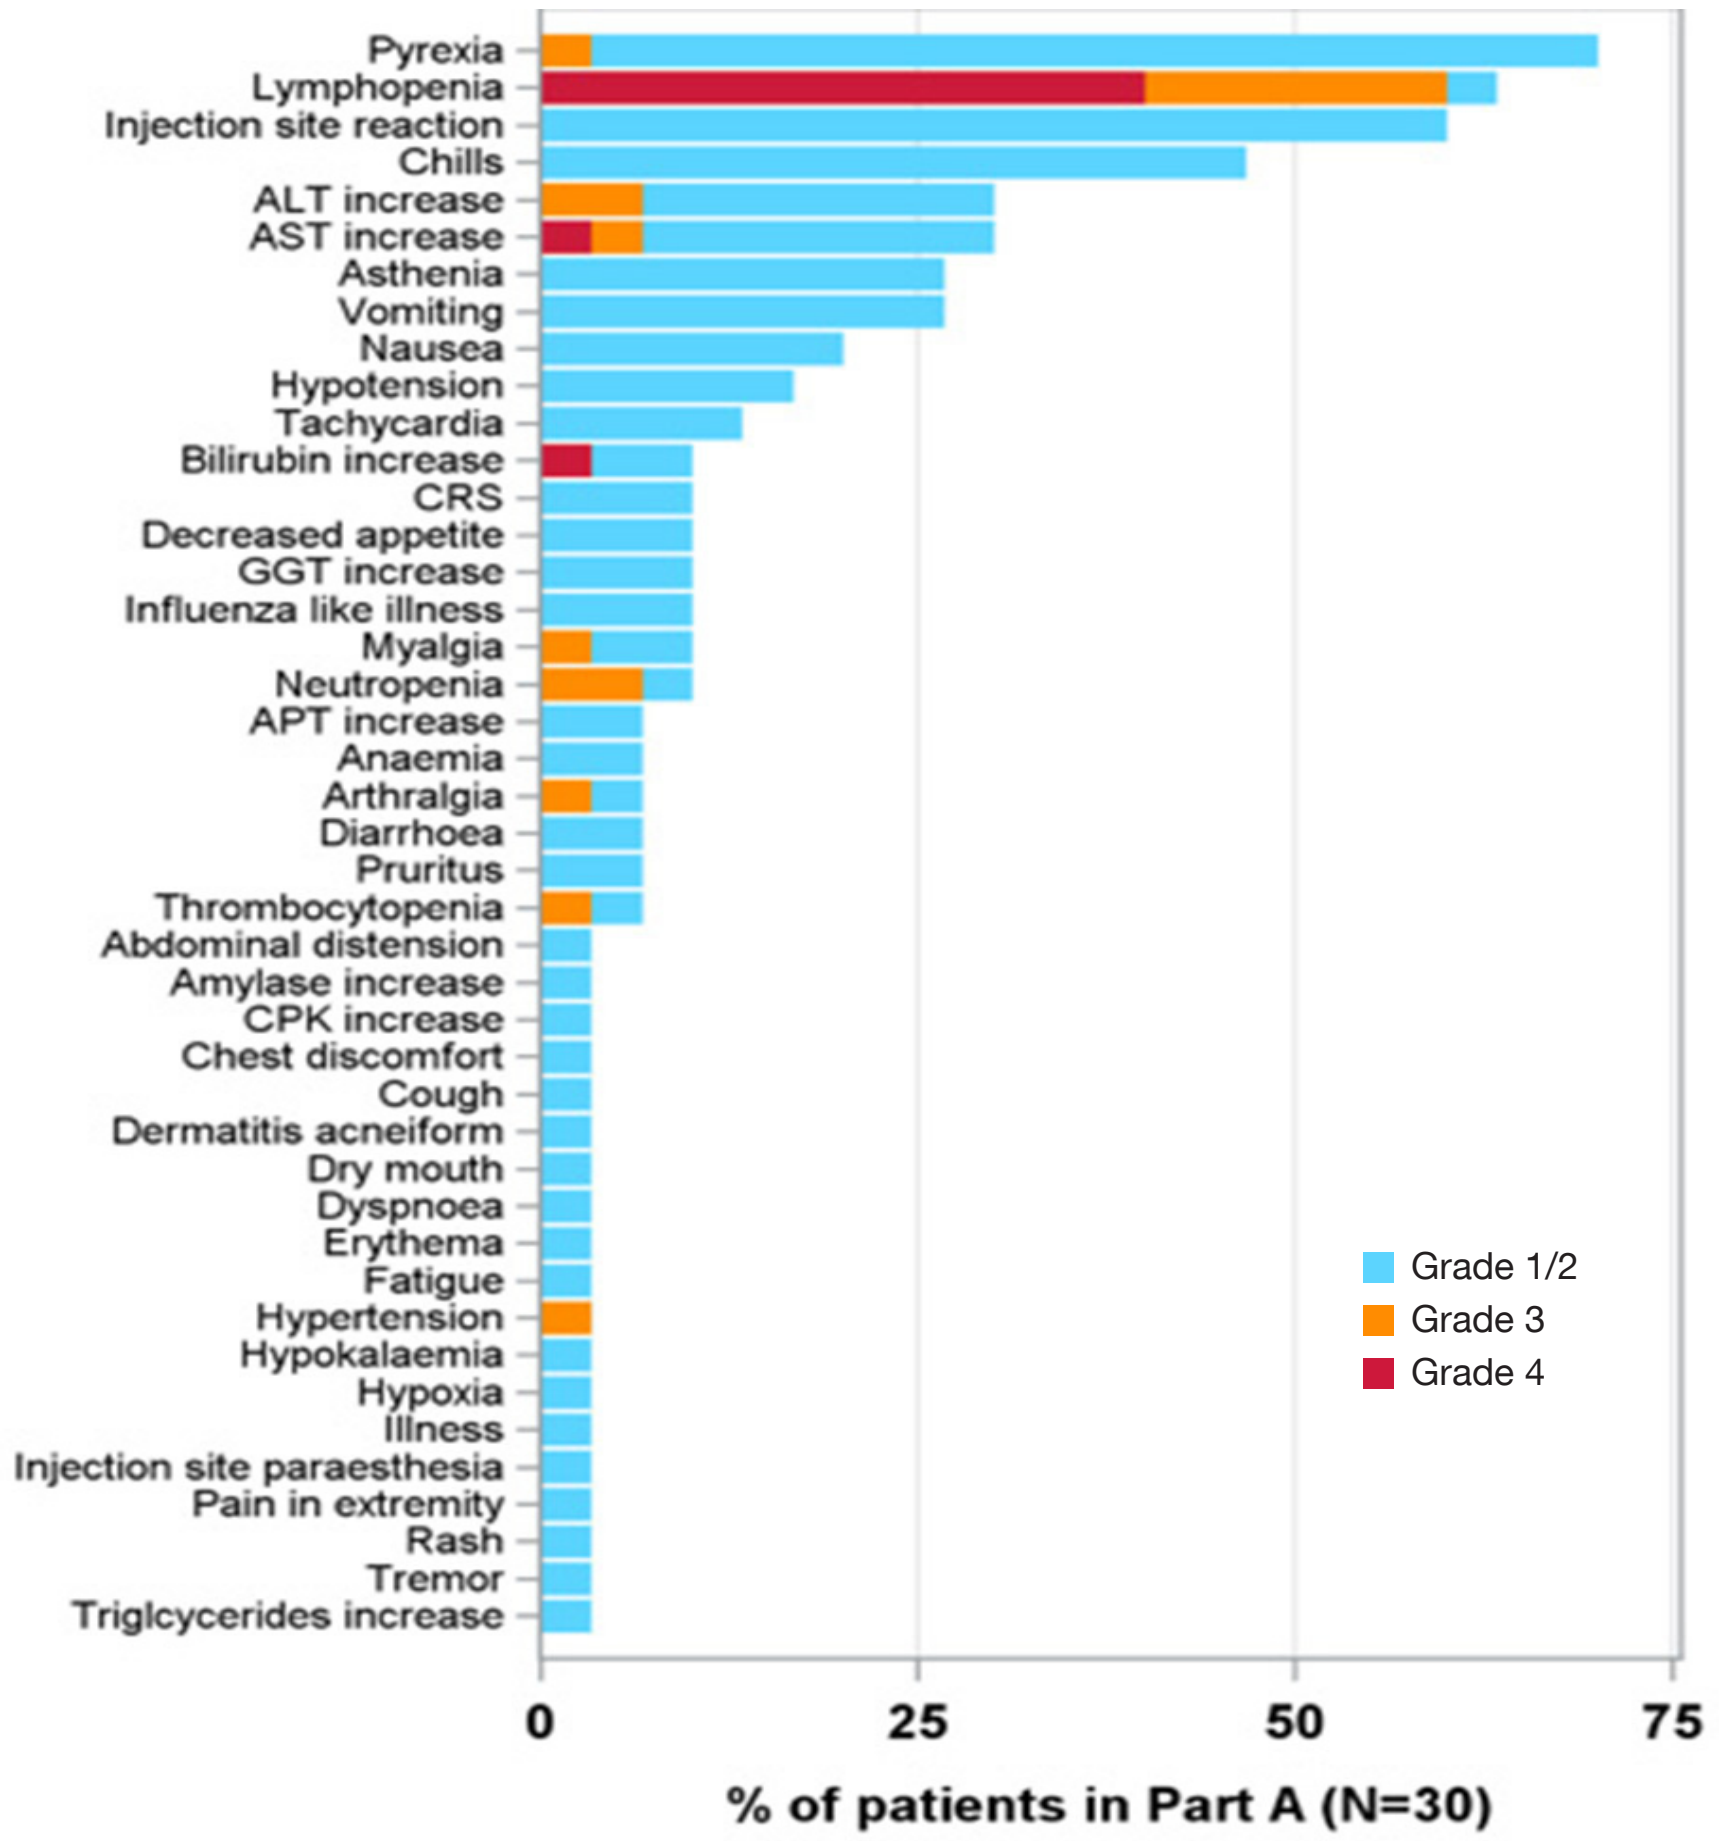
Pyrexia
Lymphopenia
Injection site reaction
Chills
ALT increase
AST increase
Asthenia
Vomiting
Nausea
Hypotension
Tachycardia
Bilirubin increase
CRS
Decreased appetite
GGT increase
Influenza like illness
Myalgia
Neutropenia
APT increase
Anaemia
Arthralgia
Diarrhoea
Pruritus
Thrombocytopenia
Abdominal distension
Amylase increase
CPK increase
Chest discomfort
Cough
Dermatitis acneiform
Dry mouth
Dyspnoea
Erythema
Fatigue
Hypertension
Hypokalaemia
Hypoxia
Illness
Injection site paraesthesia
Pain in extremity
Rash
Tremor
Triglcycerides increase
Grade 1/2
Grade 3
Grade 4
0
25
50
75
% of patients in Part A (N=30)

- Safety data from 30 patients treated at 8 dose-levels indicate that SO-C101 monotherapy is well tolerated
- The majority of AEs were fever, lymphopenia, local injection site reactions, chills, transaminase increases, flu-like symptoms as well as symptoms of cytokine release syndrome (mainly ≤ Grade 2 except for lymphopenia)
- Lymphopenia is considered mode of action-related and usually resolved within a few days.

15 µg/kg was defined as MTD :

- 2 DLTs (increased liver function tests, quickly resolved after study drug discontinuation without sequelae).

Pharmacokinetics – Cycle 1 Day 1

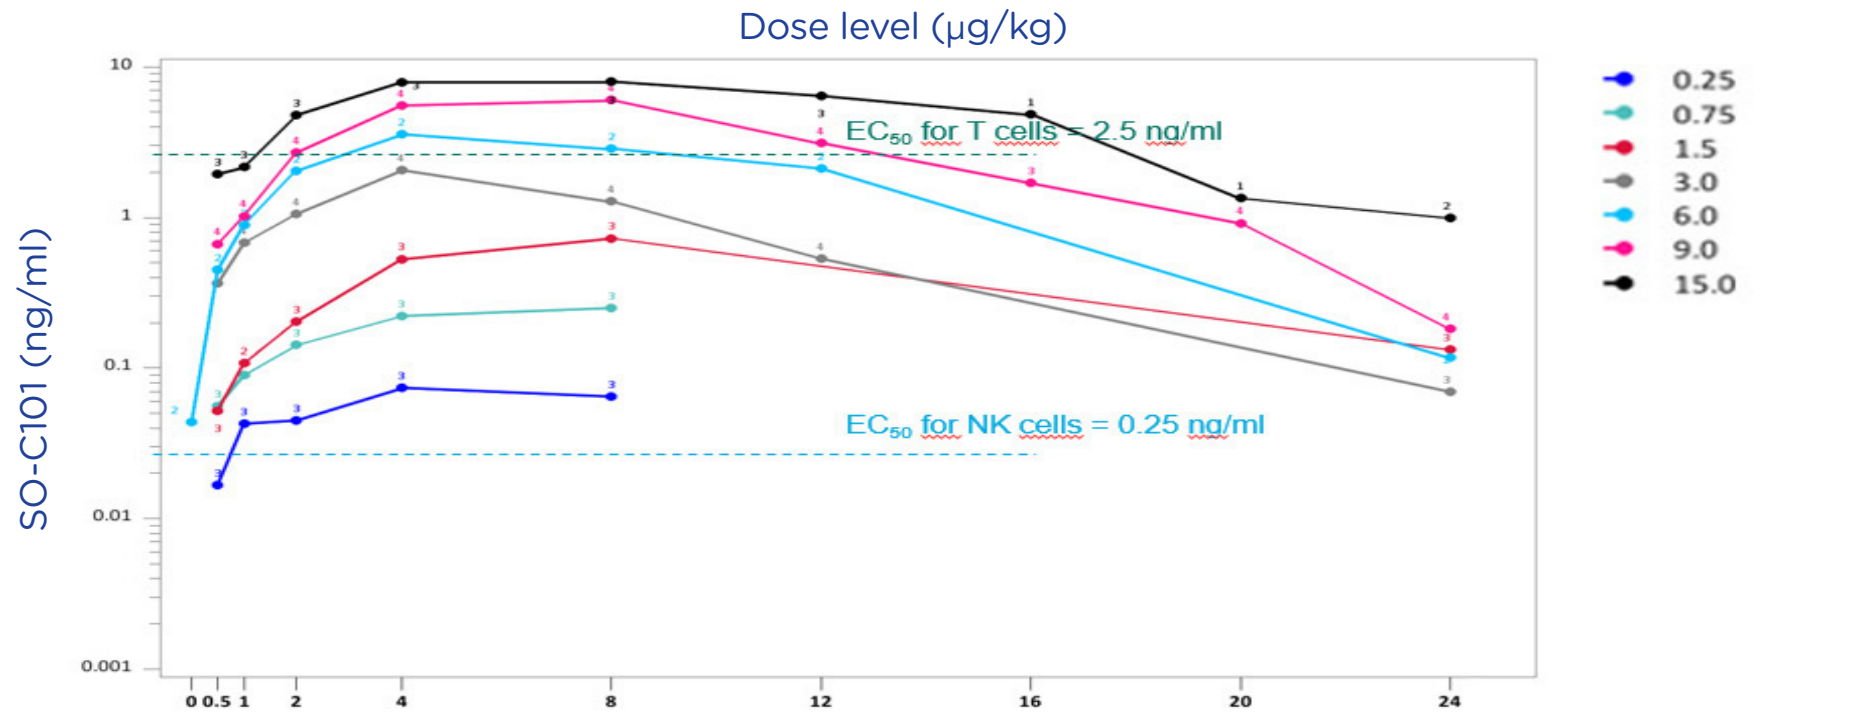
Dose level (µg/kg)
SO-C101 (ng/ml)
EC50 for T cells = 2.5 ng/ml
EC50 for NK cells = 0.25 ng/ml
0.25
0.75
1.5
3.0
6.0
9.0
15.0

- Exposure was approx. dose proportional.
- T_{max} was reached around 5 – 6 hours after administration.
- $T_{½}$ in was in the 4 hours range.

Pharmacodynamics

Maximal values of NK cell activation per dose level
Ki-67 expression %

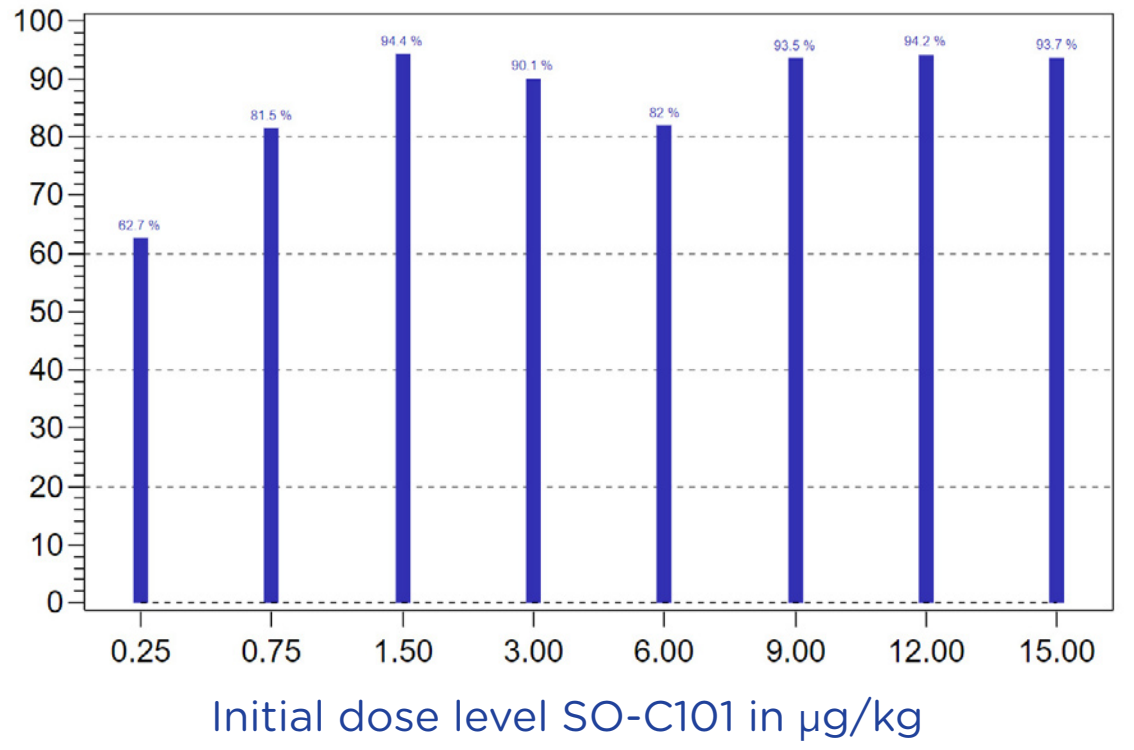
62.7 %
81.5 %
94.4 %
90.1 %
82 %
93.5 %
94.2 %
93.7 %
0.25
0.75
1.50
3.00
6.00
9.00
12.00
15.00
Initial dose level SO-C101 in µg/kg

Maximal values of CD8+ T cell activation per dose level
Ki-67 expression %

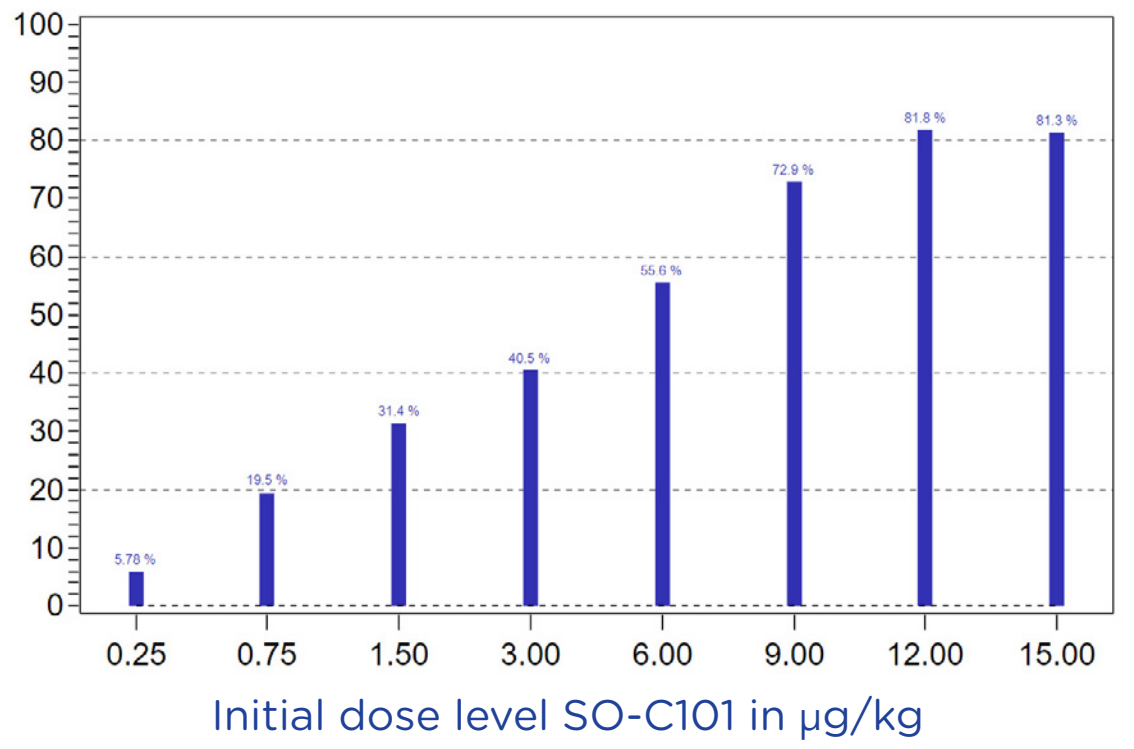
5.78 %
19.5 %
31.4 %
40.5 %
55.6 %
72.9 %
81.8 %
81.3 %
0.25
0.75
1.50
3.00
6.00
9.00
12.00
15.00
Initial dose level SO-C101 in µg/kg

- Maximum level of NK cell activation was already reached at low dose levels
- Maximum $CD8^+$ T cell activation was reached at 9 – 12 µg/kg → RP2D was selected to be 12 µg/kg

Efficacy

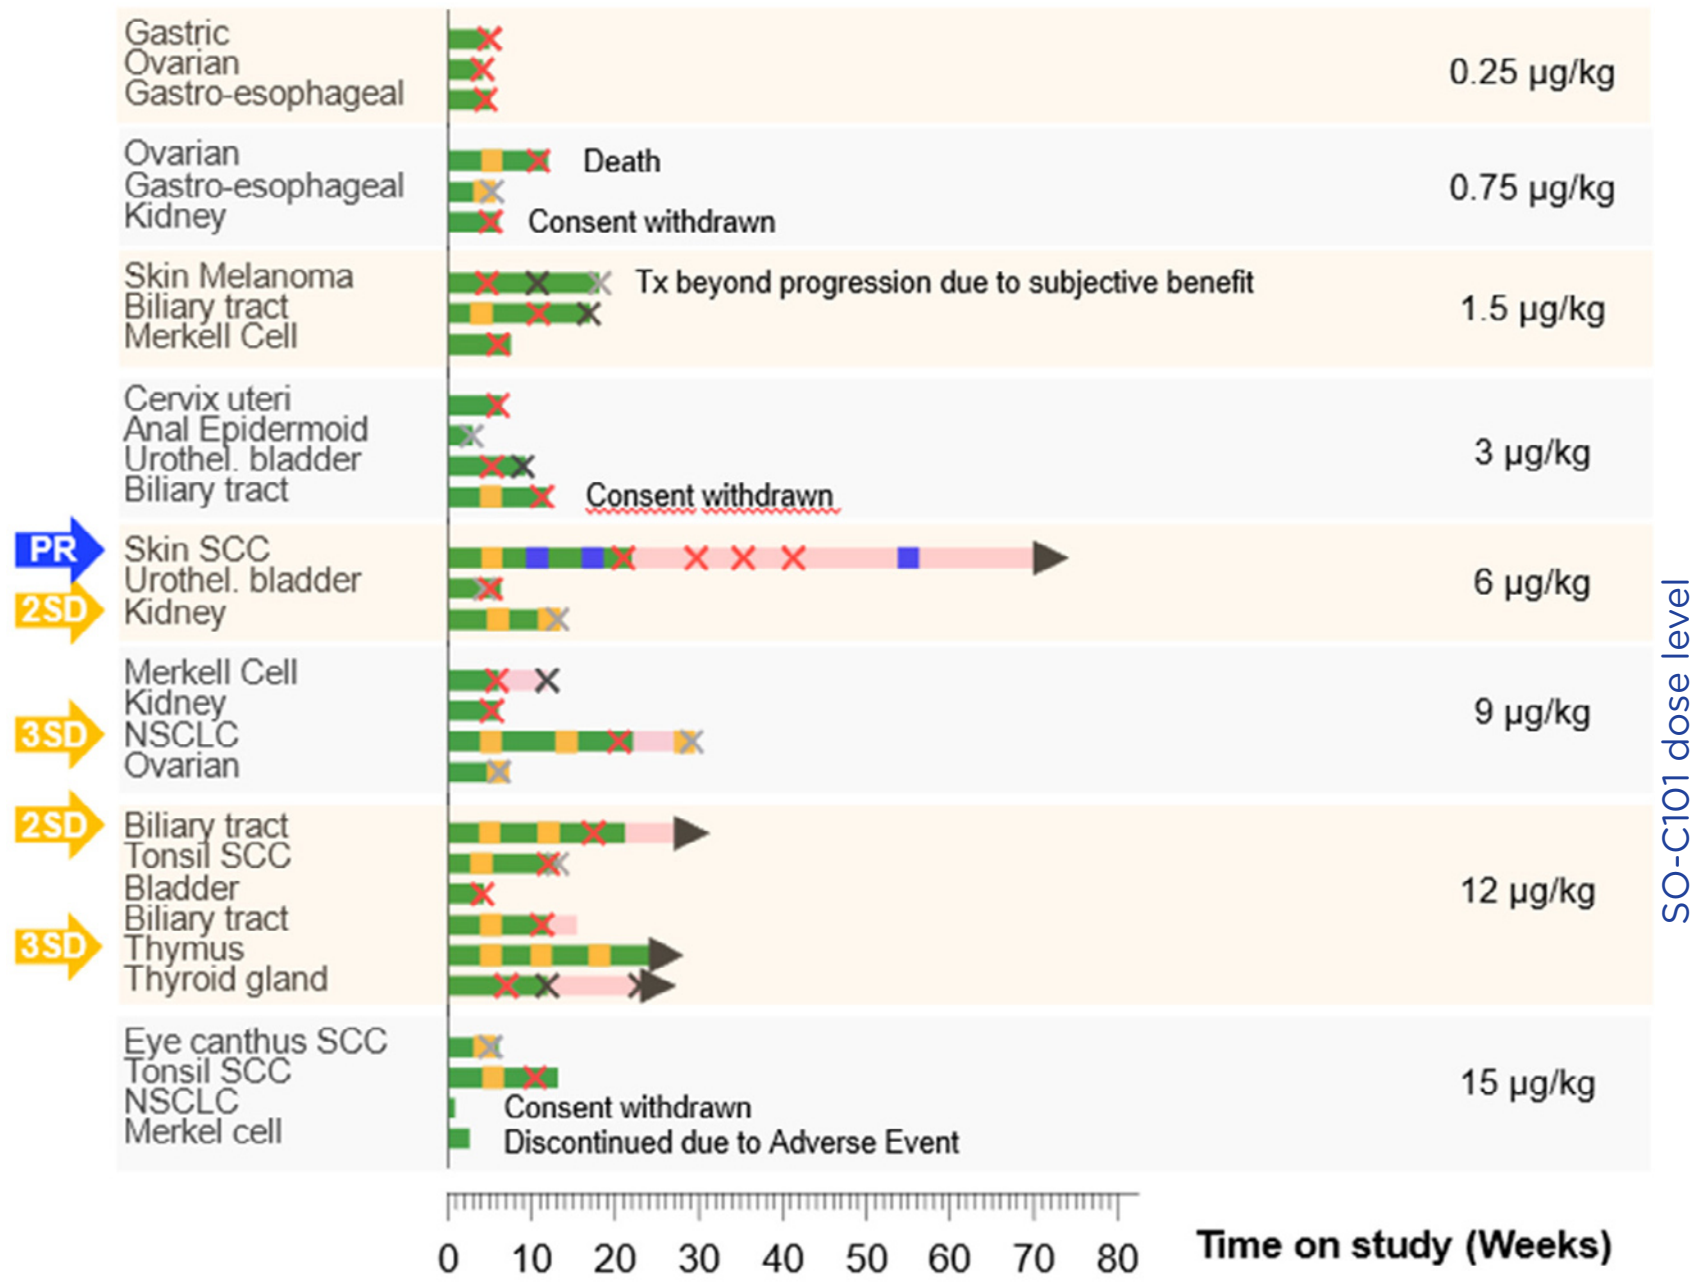
Gastric
Ovarian
Gastro-esophageal
0.25 µg/kg
Ovarian
Gastro-esophageal
Kidney
Death
Consent withdrawn
0.75 µg/kg
Skin Melanoma
Biliary tract
Merkell Cell
Tx beyond progression due to subjective benefit
1.5 µg/kg
Cervix uteri
Anal Epidermoid
Urothel. bladder
Biliary tract
Consent withdrawn
3 µg/kg
PR
2SD
Skin SCC
Urothel. bladder
Kidney
6 µg/kg
3SD
Merkell Cell
Kidney
NSCLC
Ovarian
9 µg/kg
2SD
3SD
Biliary tract
Tonsil SCC
Bladder
Biliary tract
Thymus
Thyroid gland
12 µg/kg
Eye canthus SCC
Tonsil SCC
NSCLC
Merkel cell
Consent withdrawn
Discontinued due to Adverse Event
15 µg/kg
SO-C101 dose level
0
10
20
30
40
50
60
70
80
Time on study (Weeks)

- PR in a 62 y female pt. with SSCC, previously CPI refractory
- Long-lasting SDs:
 - 71 y male pt. with Kidney cancer, 7 previous lines, CPI relapsed, SD for 188 days
 - 47 y male pt. with NSCLC, 5 previous lines, CPI relapsed, SD for 143 days
 - 57 y female pt. with Biliary tract carcinoma, 4 previous lines, CPI relapsed, SD for 148 days
 - 47 y female with Thymus carcinoma, 4 previous lines of treatment, CPI relapsed, SD for 173 days

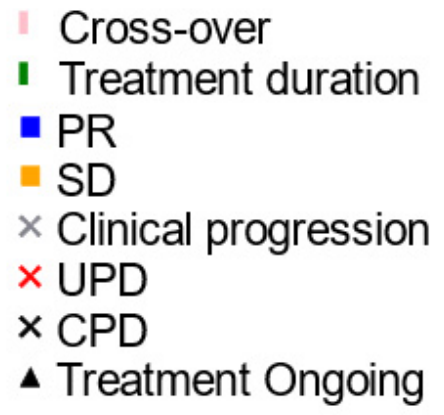
Cross-over
Treatment duration
PR
SD
Clinical progression
UPD
CPD
Treatment Ongoing

Best response of target lesions

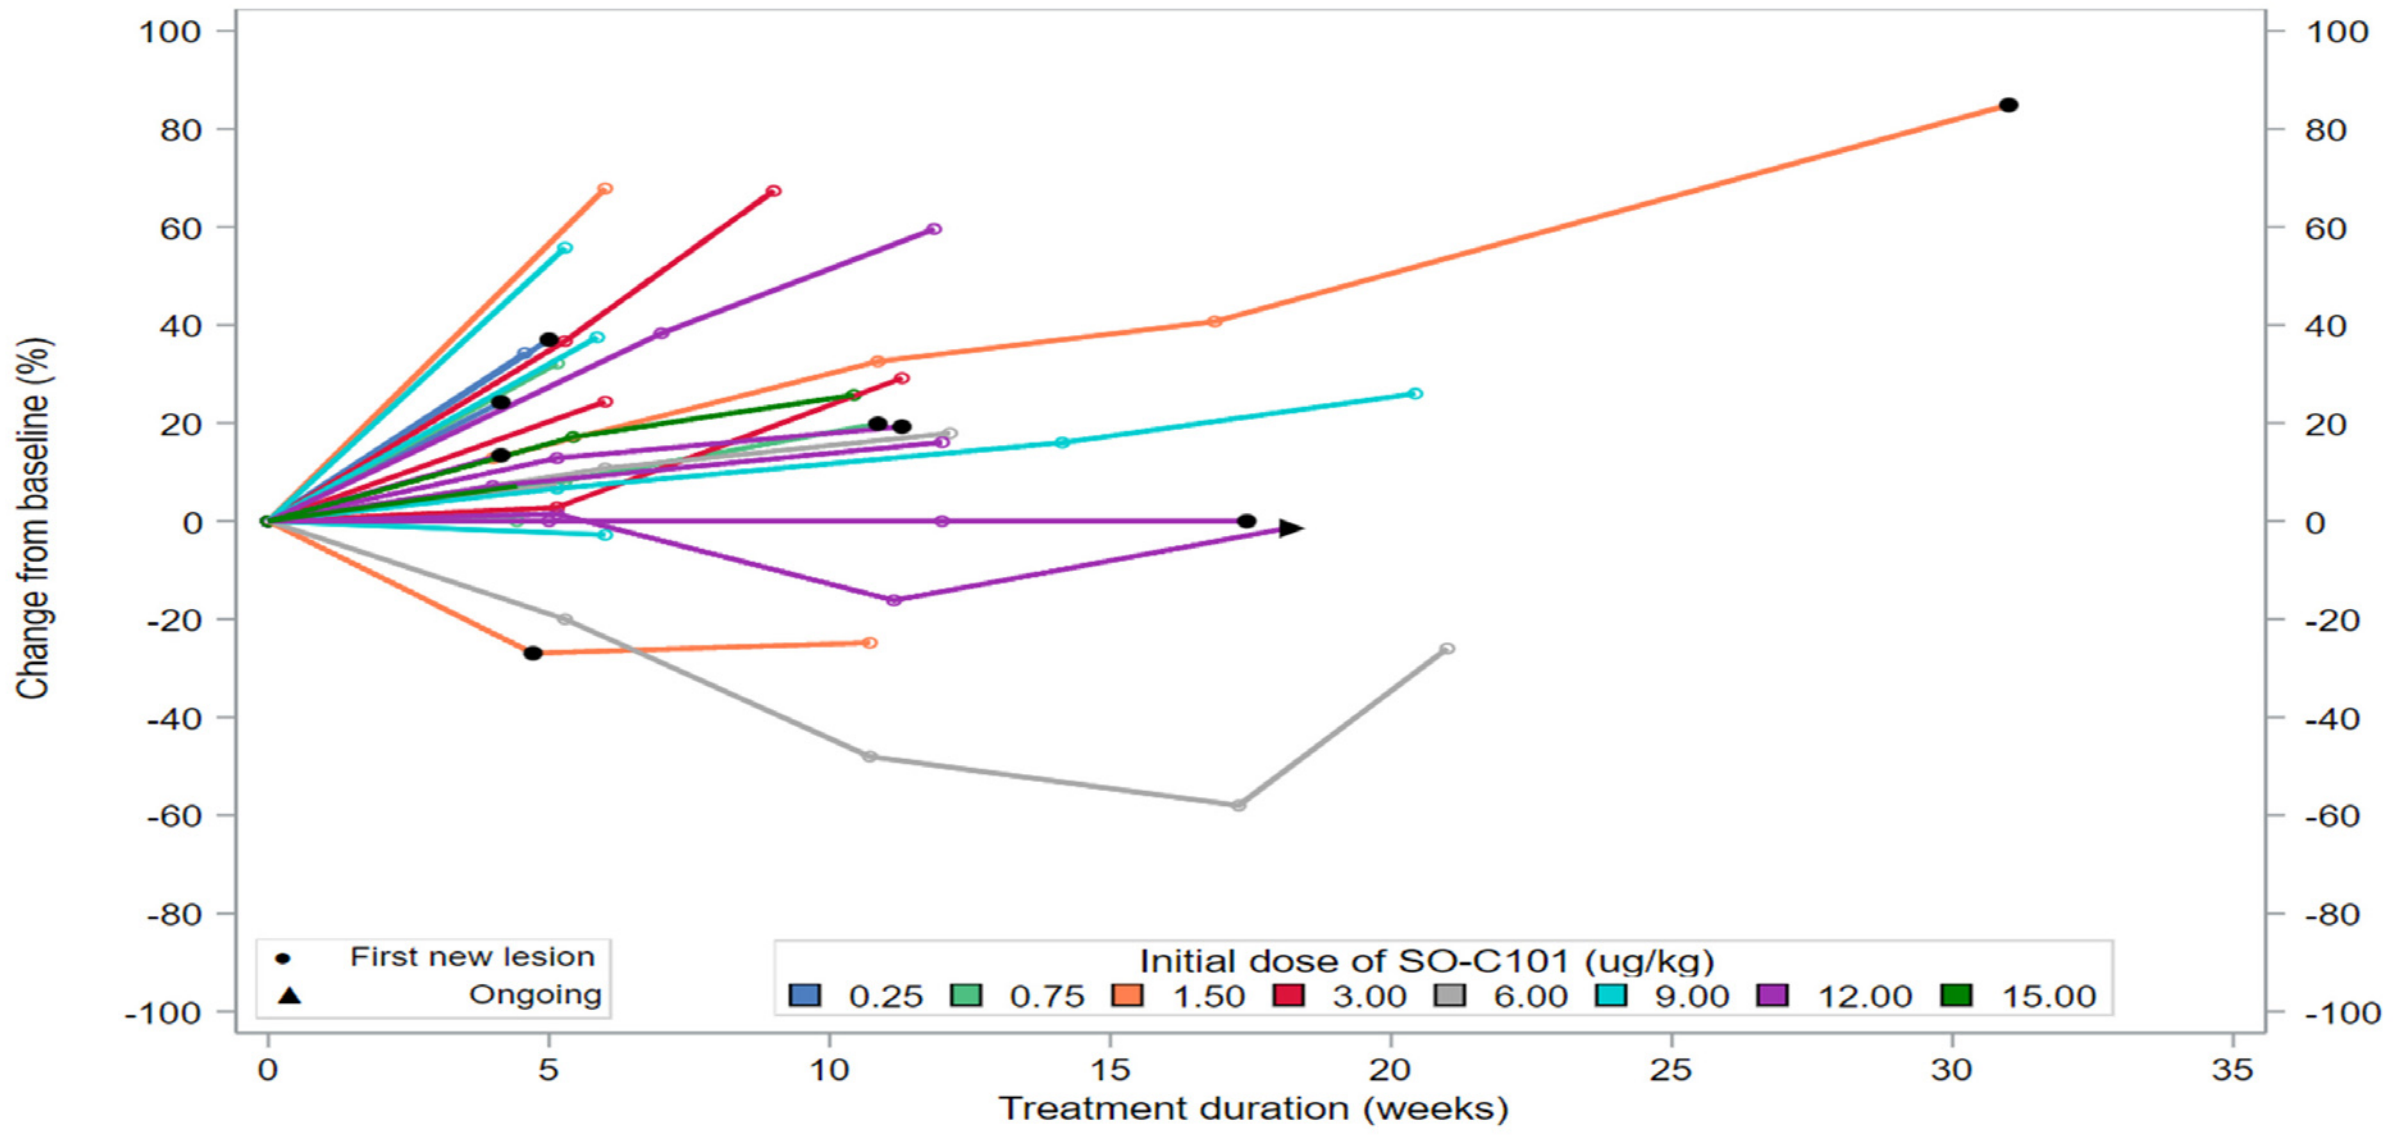
Change from baseline (%)
First new lesion
Ongoing
Initial dose of SO-C101 (ug/kg)
0.25
0.75
1.50
3.00
6.00
9.00
12.00
15.00
Treatment duration (weeks)

Response in a 62 y female patient with CPI refractory SSCC

- Previous therapies:
 - Radiation of tumour bed and lymph node at left ear (2014)
 - Docetaxel, Cisplatin, and Cetuximab (03/2019 – 06/2019)
 - **Cemiplimab** (01/2020 – 04/2020) → **refractory**

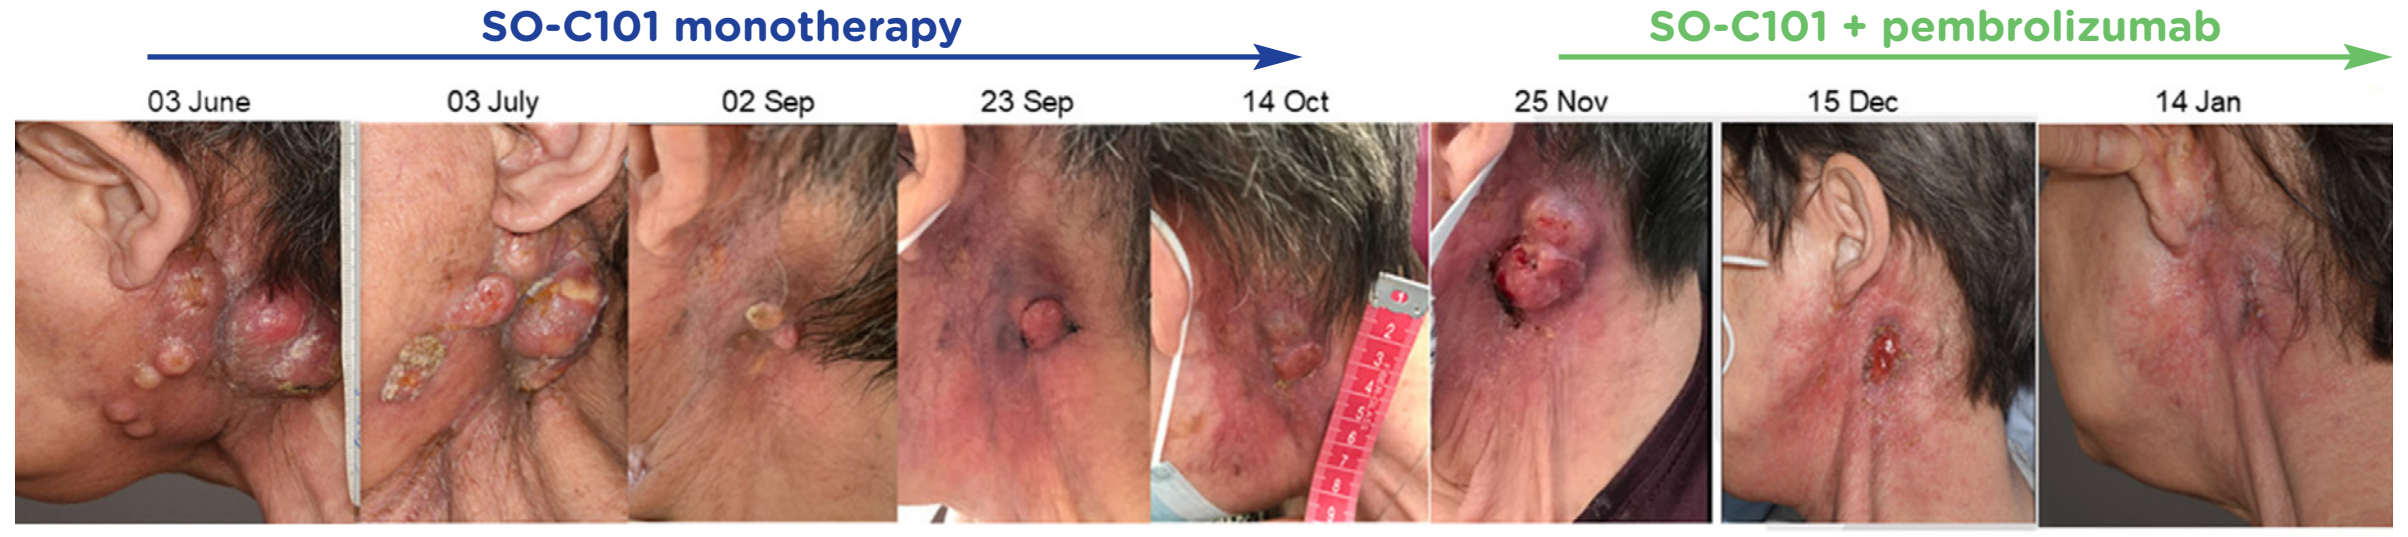
SO-C101 monotherapy
SO-C101 + pembrolizumab
03 June
03 July
02 Sep
23 Sep
14 Oct
25 Nov
15 Dec
14 Jan

SO-C101 monotherapy start: 4 June 2020

- 26 May 2020 – TL Ø 50 mm (baseline)
- 10 Jul 2020 – TL Ø 40 mm → SD (TL-20%) after 2 cycles
- 17 Aug 2020 - TL Ø 26 mm → PR (TL-49%) after 3 cycles
- 02 Oct 2020 – TL Ø 21 mm → CPR (TL- 58%) after 6 cycles
- 28 Oct 2020 – TL Ø 37 mm → PD (TL+76%) after 8 cycles → stop monotherapy due to progressive disease

SO-C1010 + Pembrolizumab start: 26 Nov 2020

- Clinical response within 2 weeks
- 28 Dec 2020 - TL Ø 19 mm; new lesion subgastric lymph node → progressive disease
- 5 May 2021: PET-CT showed no „hot spots"
- since then: TL Ø 19 mm; -62% decrease from start monotherapy, -9% from nadir on monotherapy

Conclusions

- **SO-C101 monotherapy was well tolerated**
- **The majority of TEAEs were ≤ Grade 2**
- **The RP2D was defined at 12 µg/kg**
- **Encouraging efficacy signals, even in CPI refractory patients**
 - **Clinical and radiological response in a patient with skin squamous cell carcinoma on SO-C101 monotherapy**
 - **Second response on combination therapy SO-C101+pembrolizumab after relapse on monotherapy**
 - **Additional confirmed stable disease in four patients on monotherapy**
- **A phase 2 monotherapy expansion at RP2D in selected tumour indications is ongoing**

Download the poster:

References

1. Perdreau H, et al. Eur Cytokine Netw. 2010 Dec;21(4):297-307;
2. Bessard A, et al. Mol Cancer Ther. 2009 Sep;8(9):2736-45;
3. Desbois M, et al. J Immunol. 2016 Jul 1;197(1):168-78.