Poster nr. 484

Preliminary efficacy of the IL-15 superagonist SO-C101 in combination with pembrolizumab in patients with advanced/metastatic solid tumors

Ellena Garralda[1], Vladimir Galvao[1], Stephane Champiat[2], Patricia LoRusso[3], Peter Grell[4], Aung Naing[5], Filip Janku[5], Lenka Palova Jelinkova[6], Irena Adkins[6], Ulrich Moebius[6], Richard Sachse[7], David Bechard[8], Joachim Kiemle-Kallee[7], Aurelien Marabelle[2];

SC103 Investigators: 1. Hospital Universitari Vall d'Hebron, Institute of Oncology (VHIO), Barcelona, Spain; 2. Institute Gustave Roussy Cancerologie, Villejuif, France; 3. Yale School of Medicine, New Haven, CT, United States, 4. Masaryk Memorial Cancer Institute, Brno, Czech Republic; 5. Department of Investigational Cancer Therapeutics, MD Anderson Cancer Center, Houston, TX; 6. SOTIO Biotech a.s, Prague, Czech Republic; 7. SOTIO Biotech AG, Basel, Switzerland; 8. Cytune Pharma S.A.S., Nantes, France

Introduction

- SO-C101 (future compound code: SOT101) is a superagonist fusion protein of IL-15 and the IL-15 receptor α sushi+ domain that selectively activates and expands natural killer (NK) and cytotoxic ($CD8^+$) T cells (1, 2).
- SO-C101 was shown to bind to the intermediate affinity IL-2/15 Rβγ with high affinity and specificity (1, 2).
- Synergistic effects of SO-C101 and an anti-programmed cell death protein 1 (PD-1) antibody have been validated in various tumour mouse models inducing a protective memory response (3).
- In this phase 1/1b multicentre, open-label, dose escalation study SO-C101 in combination with pembrolizumab was investigated in patients with selected advanced/metastatic tumours (NCT04234113).

Methods

- Study objectives are to determine the maximum tolerated dose (MTD), recommended phase 2 dose (RPD), pharmacokinetics, pharmacodynamics, and preliminary efficacy of SO-C101 in combination with pembrolizumab in patients with advanced/metastatic solid tumours.
- SO-C101 is administered as s.c. injection on day 1, 2, 8 and 9 of each 21 day cycle in combination with pembrolizumab i.v. 200 mg q3w.
- The evaluation period for dose-limiting toxicities is 21 days.
- The RP2D is defined as MTD or below, also considering pharmacokinetic and pharmacodynamic parameters.
- Efficacy is evaluated according to RECiST 1.1 and iRECiST.

Patient population

- Fourteen patients with a median of 2 (range 1-6) lines of previous systemic therapies were treated at dose levels 1.5, 3.0, 6.0, and 9 µg/kg BW.

Safety

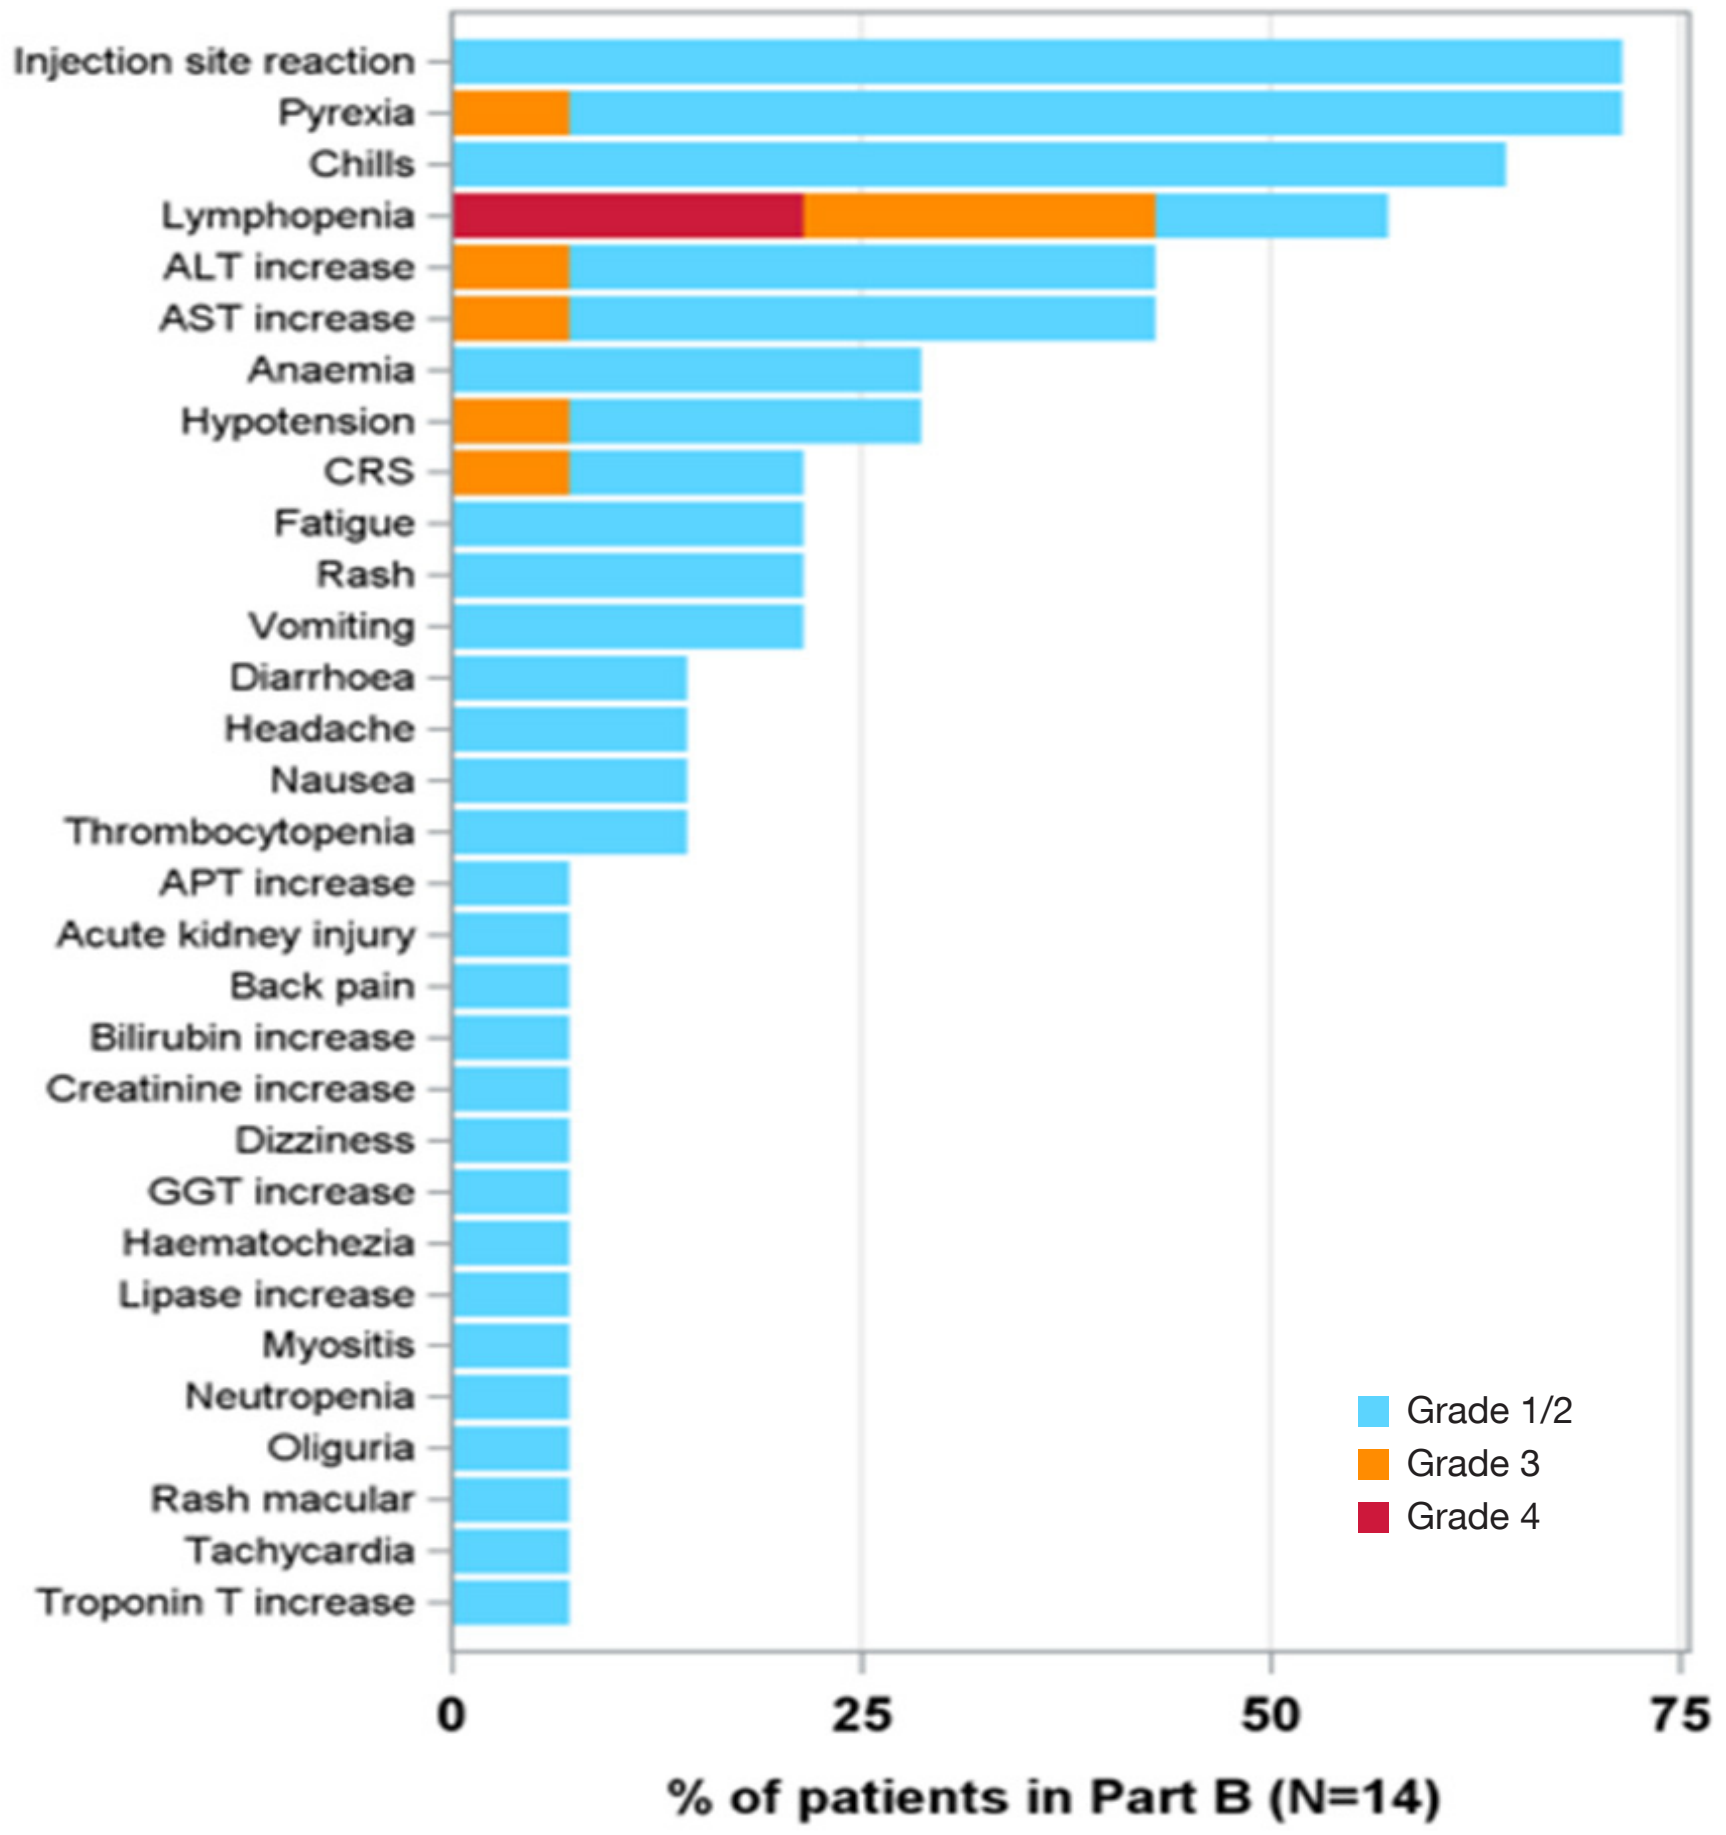
Injection site reaction
Pyrexia
Chills
Lymphopenia
ALT increase
AST increase
Anaemia
Hypotension
CRS
Fatigue
Rash
Vomiting
Diarrhoea
Headache
Nausea
Thrombocytopenia
APT increase
Acute kidney injury
Back pain
Bilirubin increase
Creatinine increase
Dizziness
GGT increase
Haematochezia
Lipase increase
Myositis
Neutropenia
Oliguria
Rash macular
Tachycardia
Troponin T increase
Grade 1/2
Grade 3
Grade 4
0
25
50
75
% of patients in Part B (N=14)

- SO-C101 in combination with pembrolizumab was well tolerated.
- The adverse event profile was consistent with the monotherapy AE profile from either single agent compound.
- Dose level 6 µg/kg expanded to 7 patients due to a DLT
 - CRS grade 3 (DLT) in one patient after first administration. The patient continued the study on a reduced dose (3 µg/kg).
- Dose level 9 µg/kg is ongoing.

SO-C101 in combination with Pembrolizumab induces partial response and long-lasting stable disease

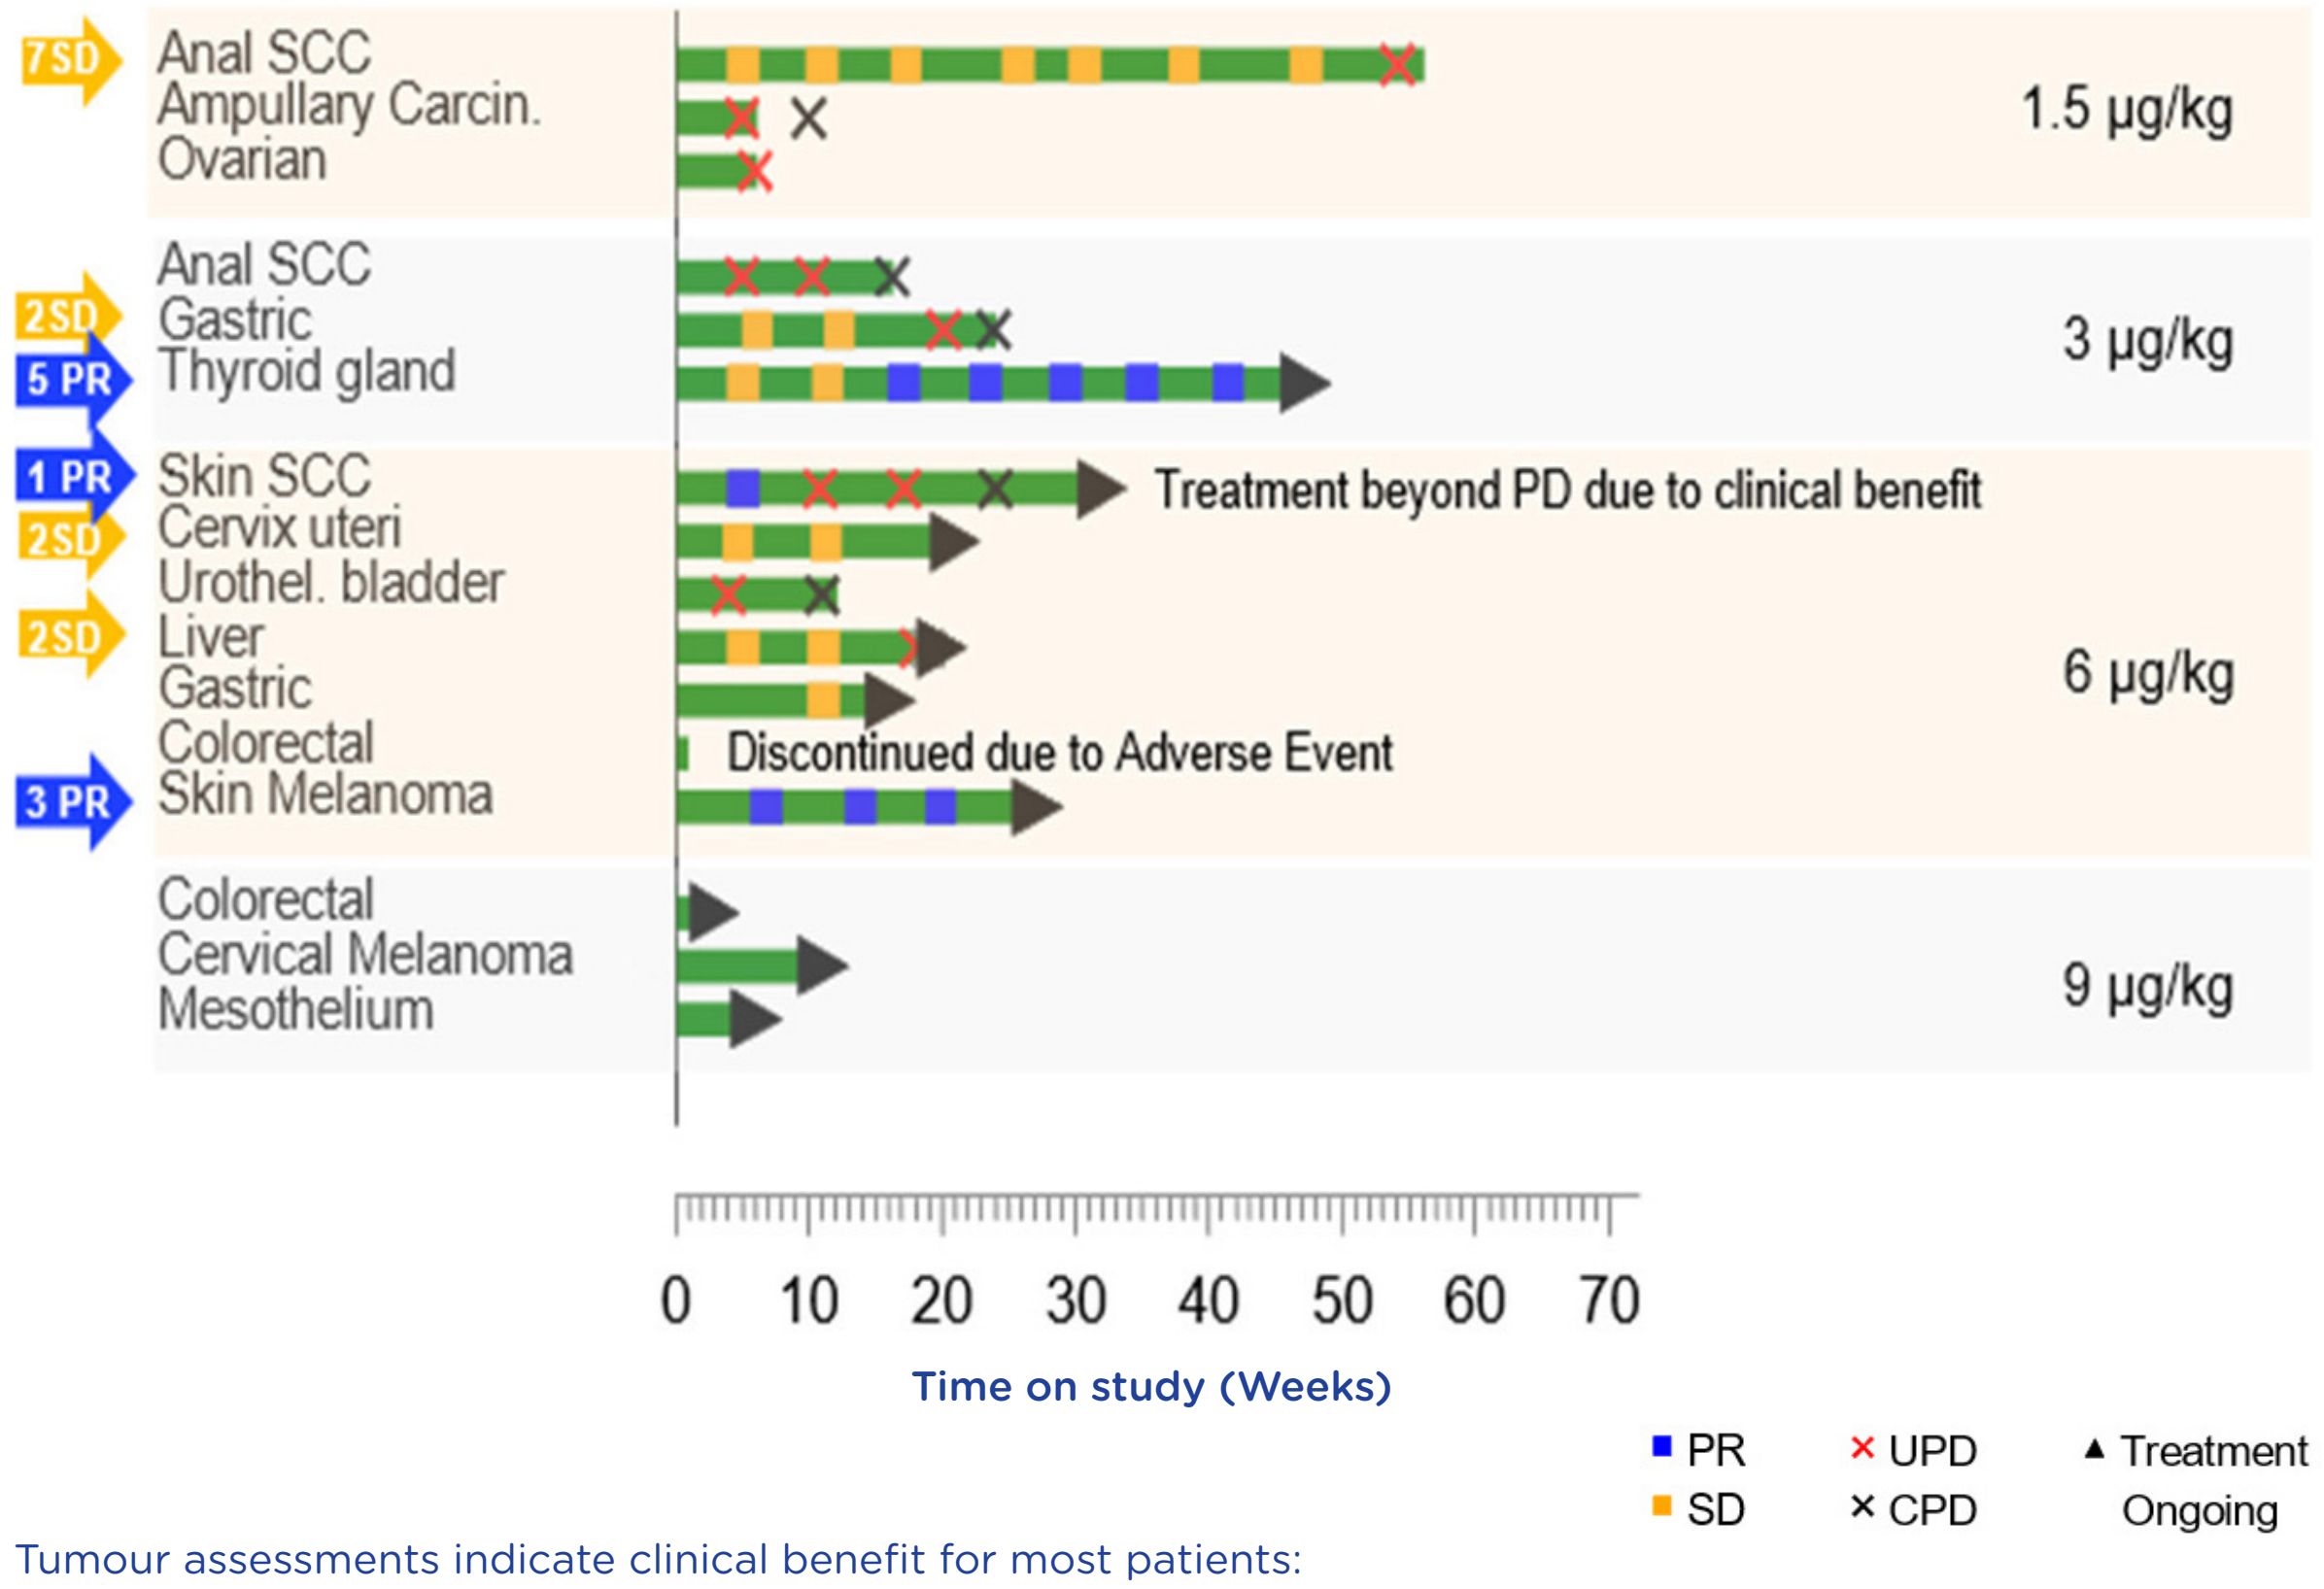
7SD
Anal SCC
Ampullary Carcin.
Ovarian
1.5 µg/kg
2SD
5 PR
Anal SCC
Gastric
Thyroid gland
3 µg/kg
1 PR
2SD
2SD
3 PR
Skin SCC
Cervix uteri
Urothel. bladder
Liver
Gastric
Colorectal
Skin Melanoma
Treatment beyond PD due to clinical benefit
Discontinued due to Adverse Event
6 µg/kg
Colorectal
Cervical Melanoma
Mesothelium
9 µg/kg
0 10 20 30 40 50 60 70
Time on study (Weeks)
PR
SD
UPD
CPD
Treatment Ongoing

Tumour assessments indicate clinical benefit for most patients:
- Partial response in 3 patients
- Long-term stable disease in 4 patients
 - 49 y female pt. with Anal SCC, 2 previous lines, CPI relapsed, SD for 379 days
 - 64 y male pt. with Gastric cancer, 2 previous lines, SD for 141 days
 - 63 y female pt. with Cervix uteri cancer, 3 previous lines, SD since 140 days ongoing
 - 67 y female pt. with Liver cancer, 4 previous lines, CPI relapsed, SD for 133 days, ongoing

Partial Response in three patients

47 y female with thyroid gland carcinoma

- Initial diagnosis: 2008
- Previous therapies
 - Surgeries
 - Radiotherapy
 - Systemic therapy
 - Vandetanib (2014-2018)
 - no previous CPI
- Partial response
 - target lesion -36% after 10 cycles
- Status
 - Patient on treatment in cycle 15

74 y female with SSCC

- Initial diagnosis: 2016
- Previous therapies
 - 22 surgeries (2006-2020)
 - Systemic therapy
 - Cemiplimab (4 cycles) → primary resistance
- Response
 - target lesion -100% after 4 cycles
 - fluctuating new lesions coming and disappearing
- Status
 - Significant clinical response
 - Patient on treatment in cycle 7

57 y male with metastatic melanoma

- Initial diagnosis: 2018
- Previous therapies
 - Surgeries (2018)
 - Systemic therapy
 - Spartalizumab, dabrafenib and trametinib (2018 – 2021) → relapse
- Response
 - target lesion -60% after 5 cycles
- Status
 - Patient on treatment in cycle 6

Best response assessment shows benefit in majority of patients

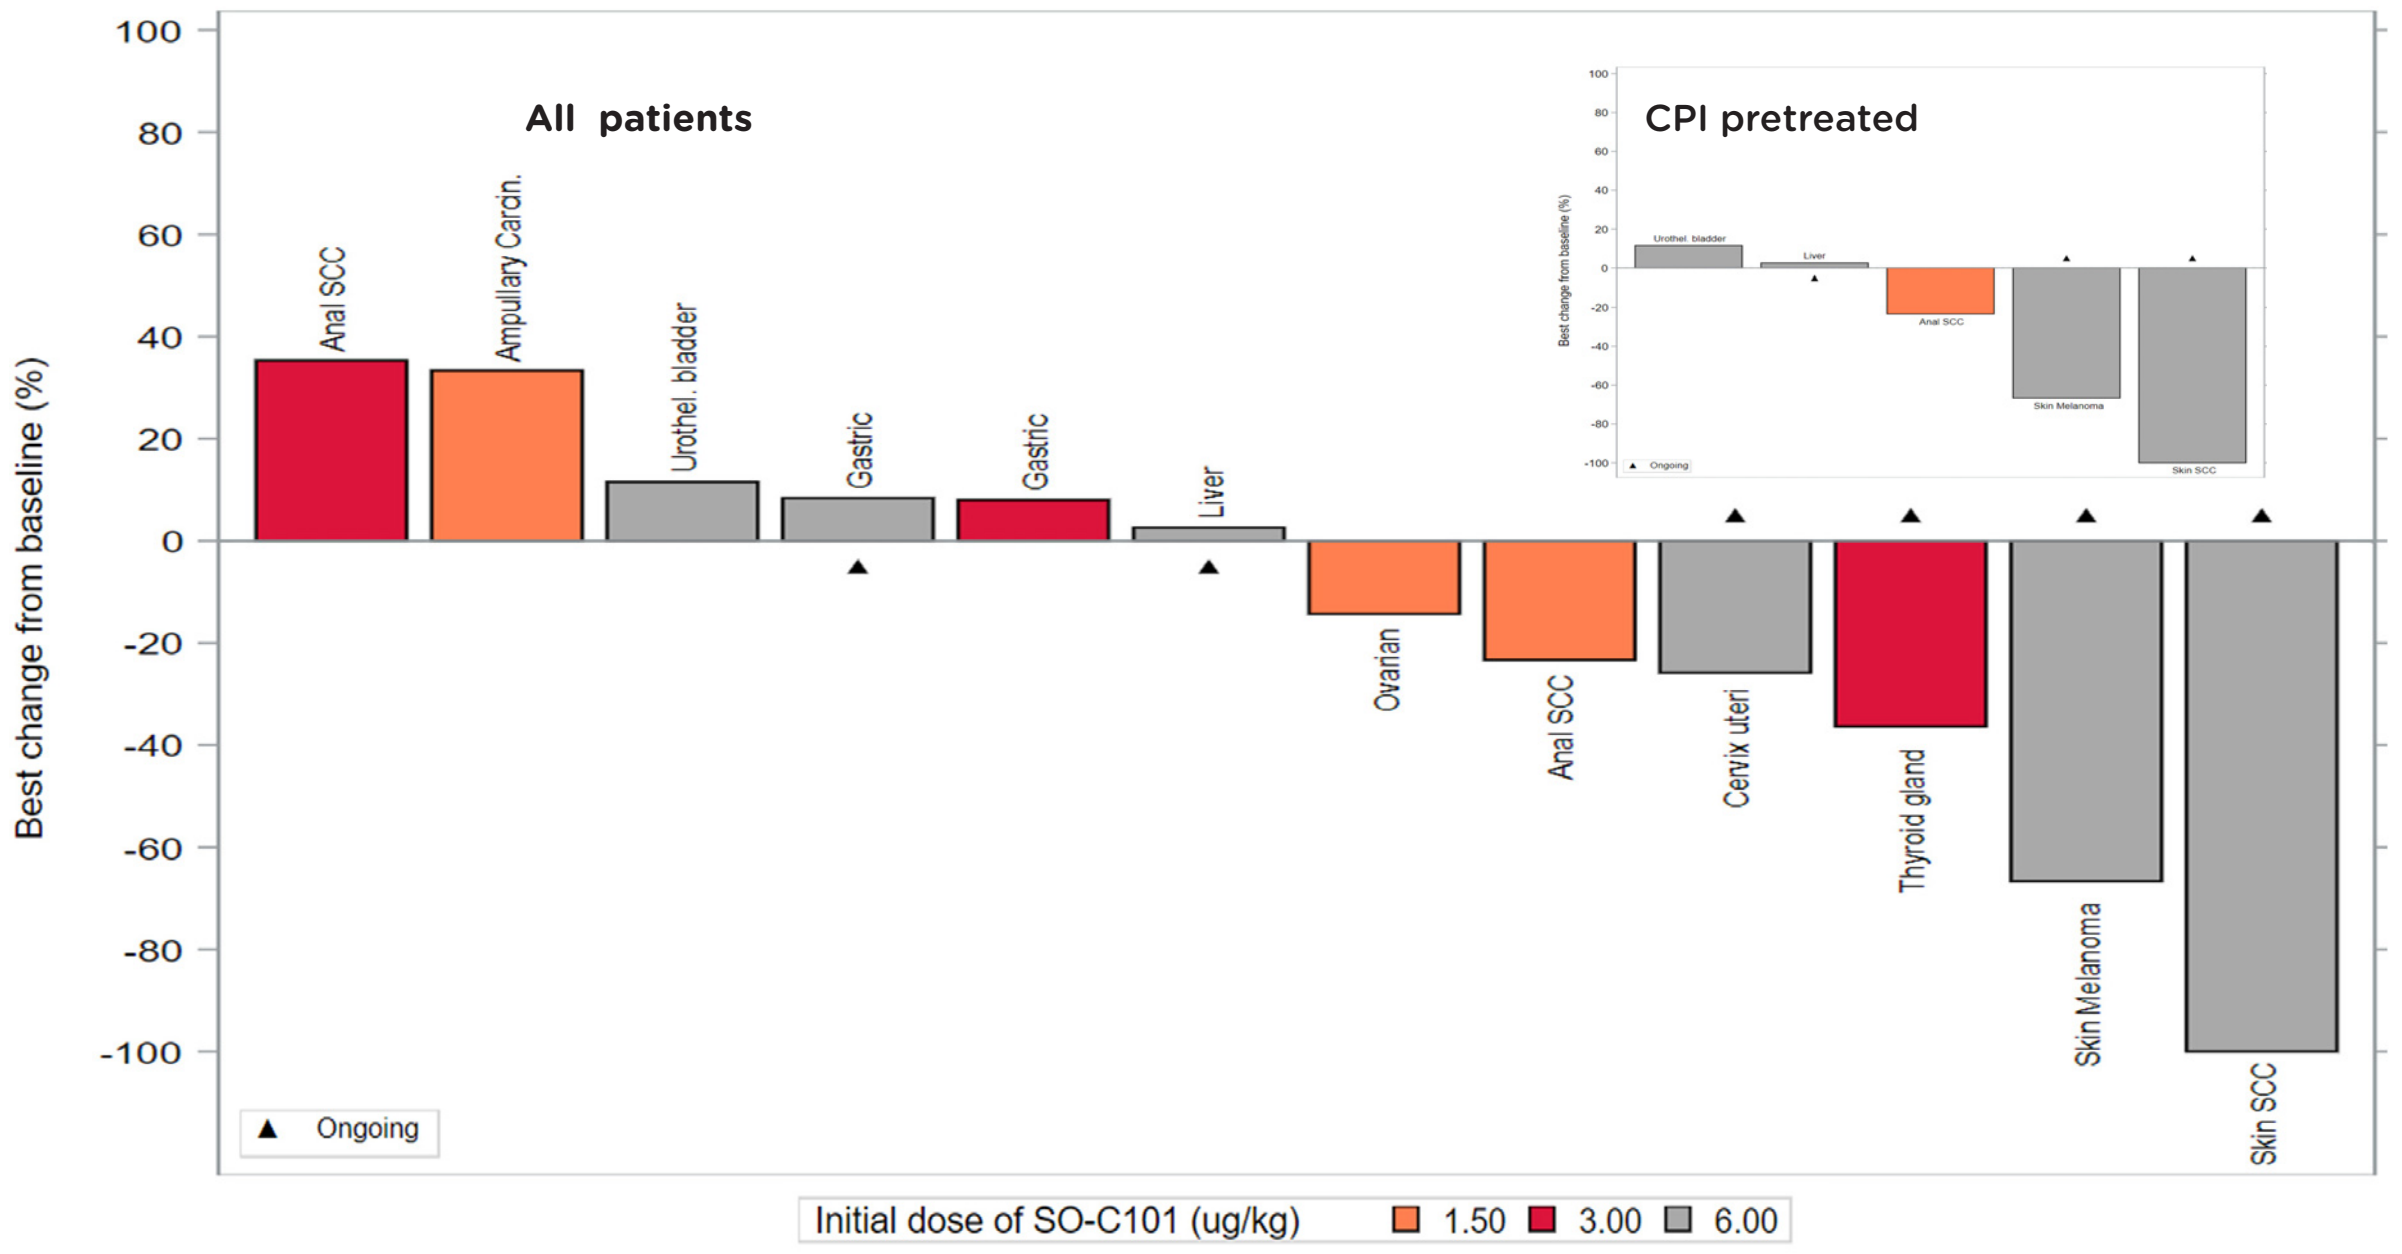
All patients
CPI pretreated
Best change from baseline (%)
Anal SCC
Ampullary Carcin.
Urothel. bladder
Gastric
Gastric
Liver
Ovarian
Anal SCC
Cervix uteri
Thyroid gland
Skin Melanoma
Skin SCC
Ongoing
Initial dose of SO-C101 (ug/kg) 1.50 3.00 6.00

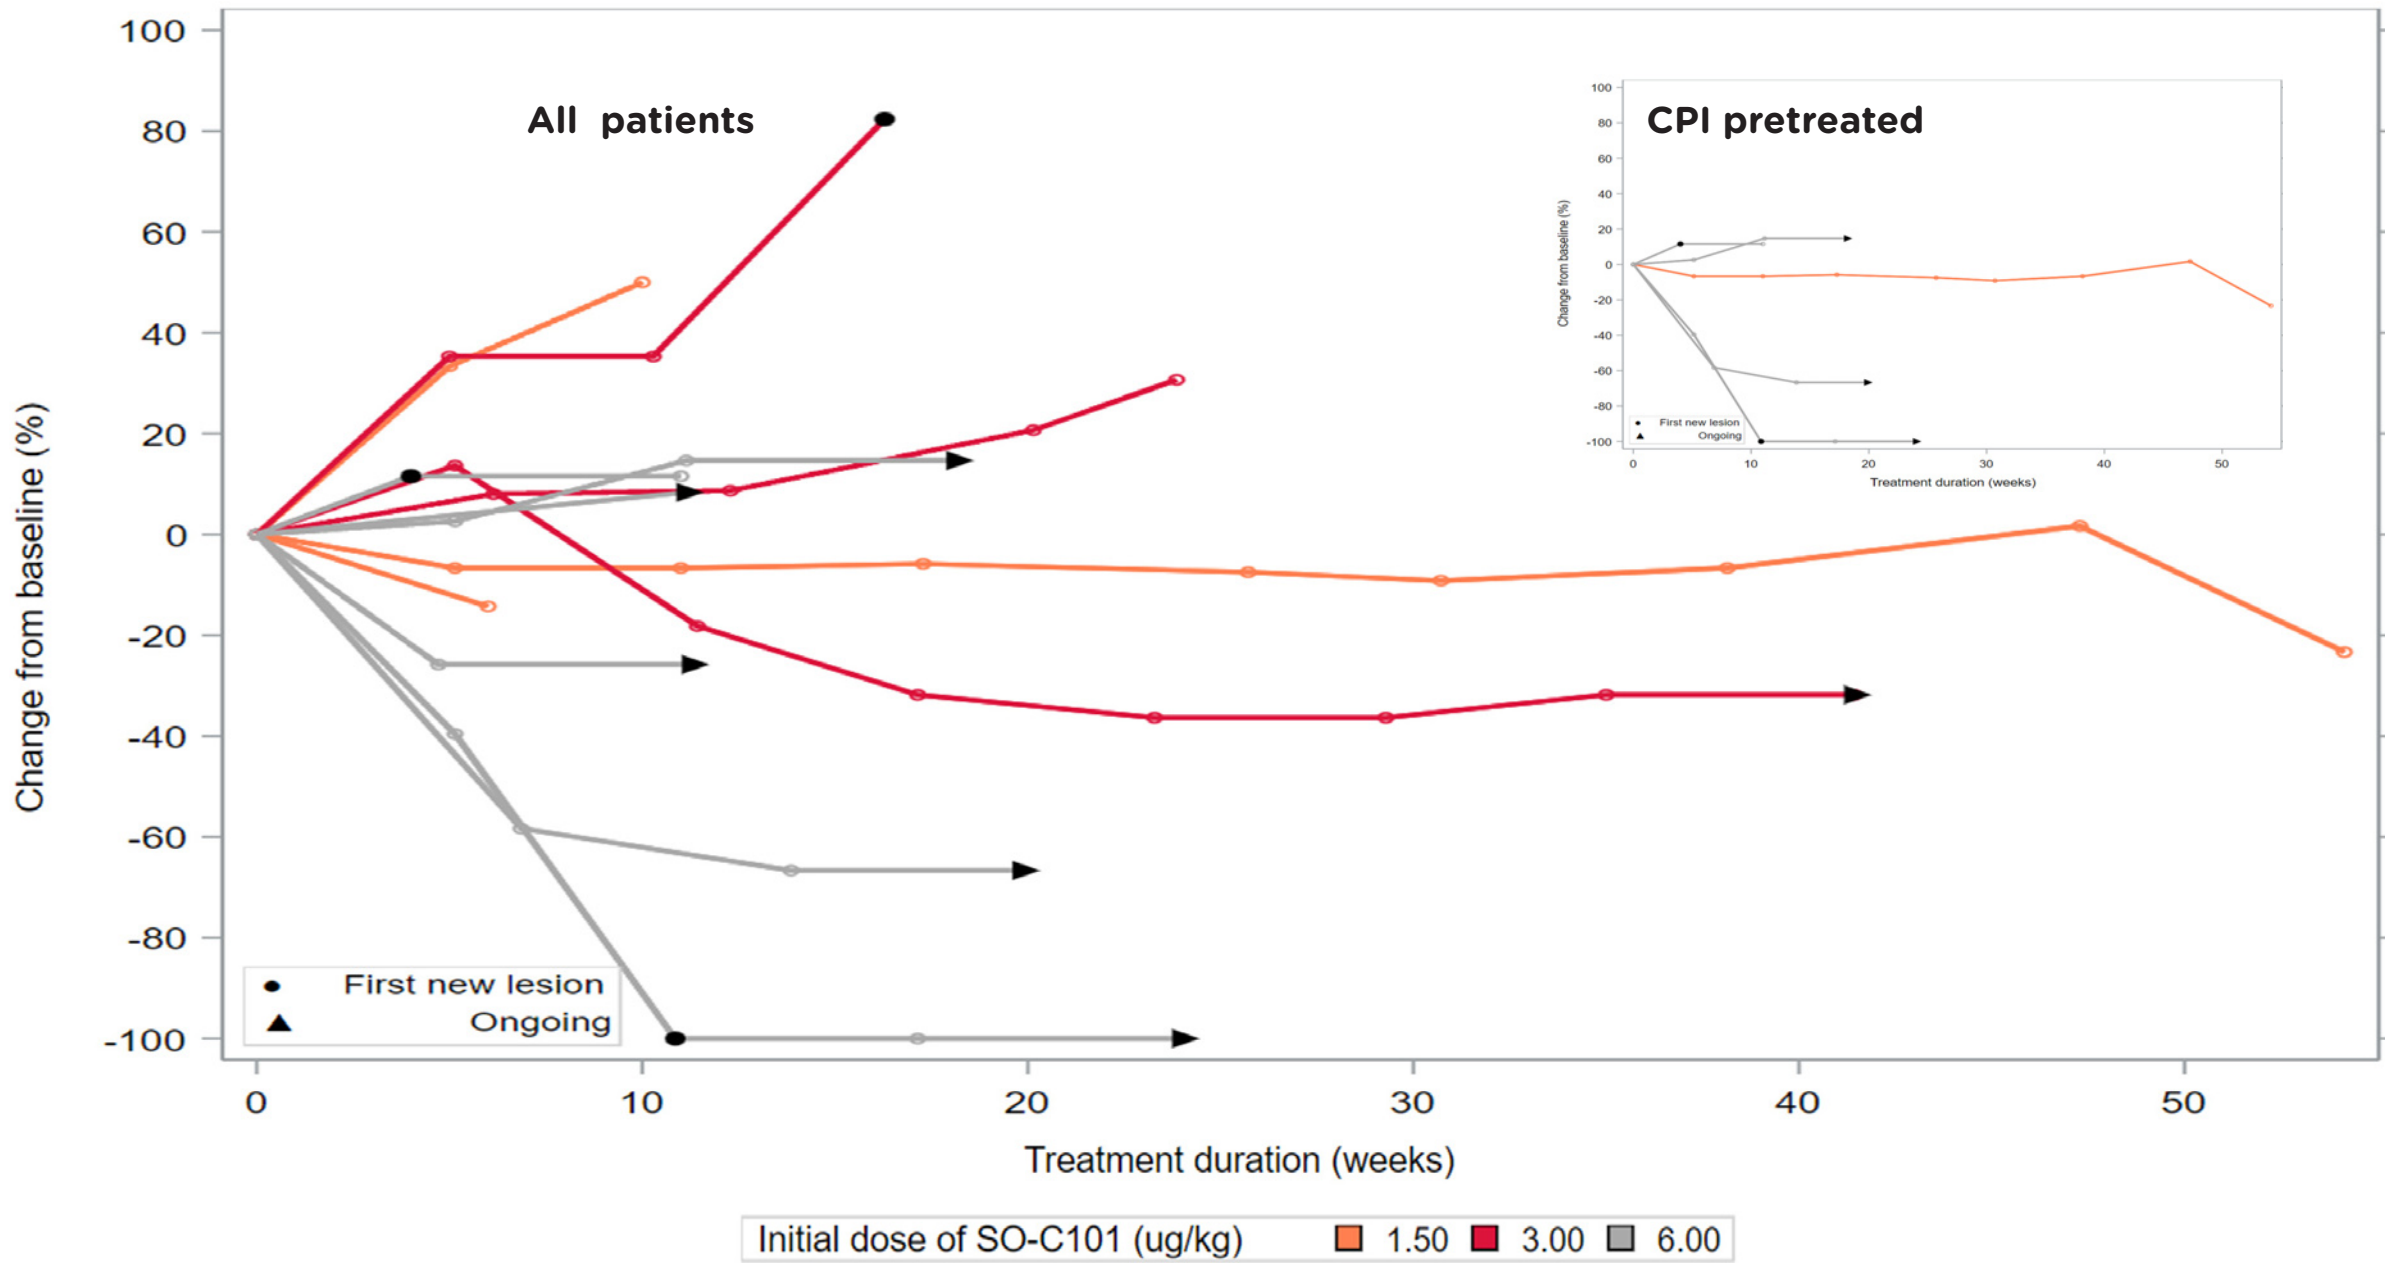
All patients
CPI pretreated
Change from baseline (%)
First new lesion
Ongoing
Treatment duration (weeks)
Initial dose of SO-C101 (ug/kg) 1.50 3.00 6.00

Conclusions

- **SO-C101 in combination with pembrolizumab was well tolerated**
- **The dose escalation is ongoing**
- **Clinical benefit observed in majority of patients, even in CPI relapsed/refractory:**
 - **Partial response in 3 patients**
 - **Long lasting stable disease in 4 patients**
- **A phase 2 basket study in selected indications is in preparation**

Download the poster:

References

1. Perdreau H, et al. Eur Cytokine Netw. 2010 ; Dec;21(4):297-307
2. Bessard A, et al. Mol Cancer Ther. 2009 Sep;8(9):2736-45
3. Desbois M, et al. J Immunol. 2016 Jul 1;197(1):168-78.